NTS-105 Decreases Cell Death After *In Vitro* Stretch Injury

Mary Kate R Dwyer, Carolyn Kim, Nevin Varghese, Barclay Morrison III

Columbia University, New York, NY

Correspondence: mrd2184@columbia.edu

COLUMBIA | ENGINEERING
The Fu Foundation School of Engineering and Applied Science

NeuroTrauma™ SCIENCES, LLC

Background

- Upwards of 280,000 people are hospitalized for traumatic brain injury (TBI) every year [1].
- *In vitro* models of TBI, such as biaxial stretch of organotypic hippocampal slices (OHSCs), allow more rapid testing of treatments for TBI. OHSCs preserve the structure, function, and cell types of the brain in order to study mechanisms of injury and treatment.
- All clinical trials for TBI have failed, including several progesterone studies, even though previous studies with progesterone showed promising results [2].
- Work is being done to leverage the potential benefits of progesterone treatments without the drawbacks.
- NTS-105 is a novel neuroactive steroid that is being tested in TBI and stroke models. It may protect against inflammation and hypoxia and is blood brain barrier permeable.

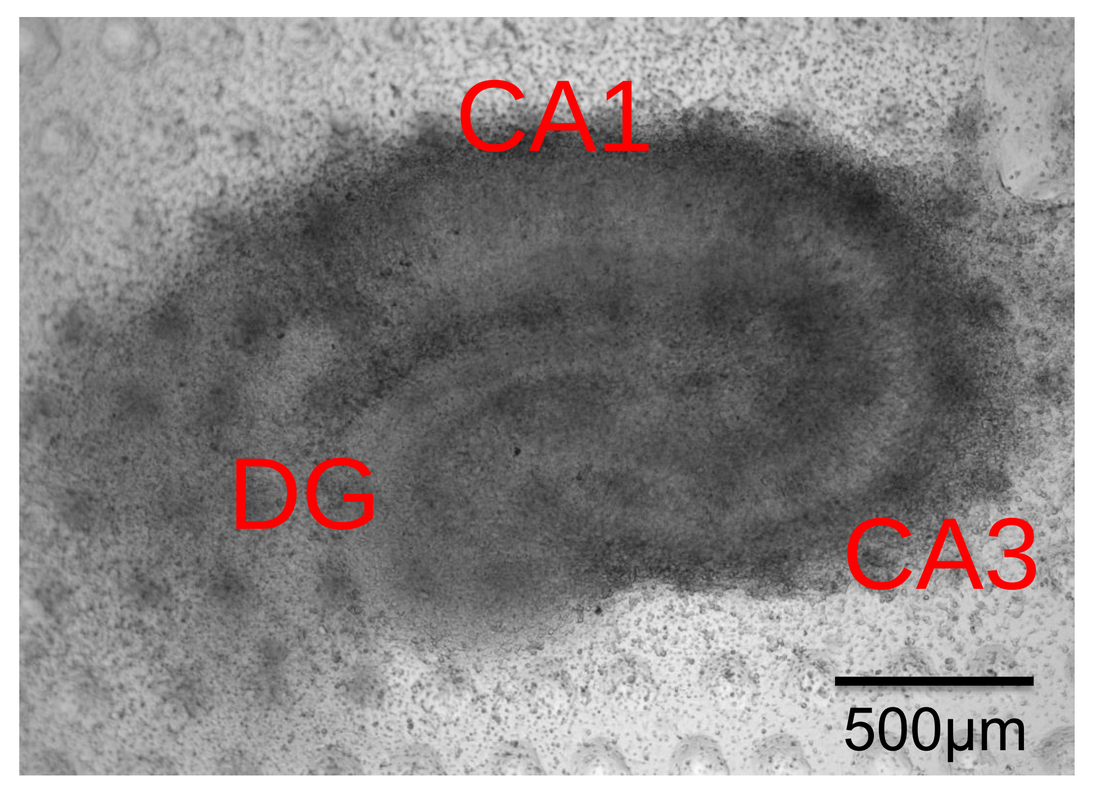
CA1
DG
CA3
500µm

Image of an OHSC on a patterned silicone membrane 10 days after culture. Hippocampal structure is maintained.

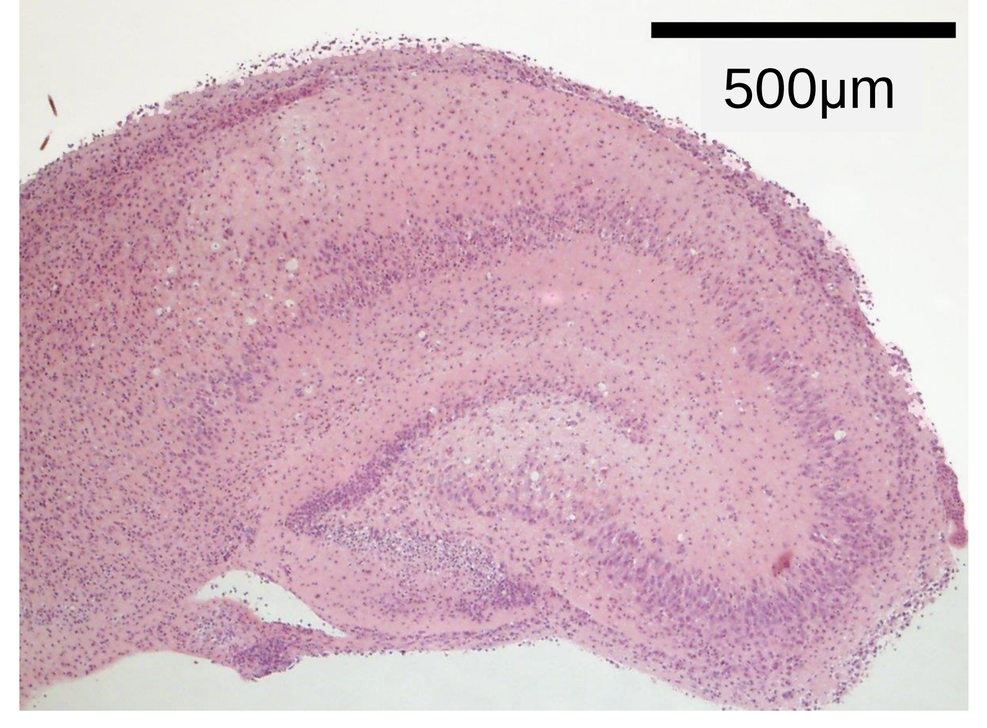
500µm

H&E stain of OHSC grown on a silicone membrane. Image reproduced from [3].

Methods

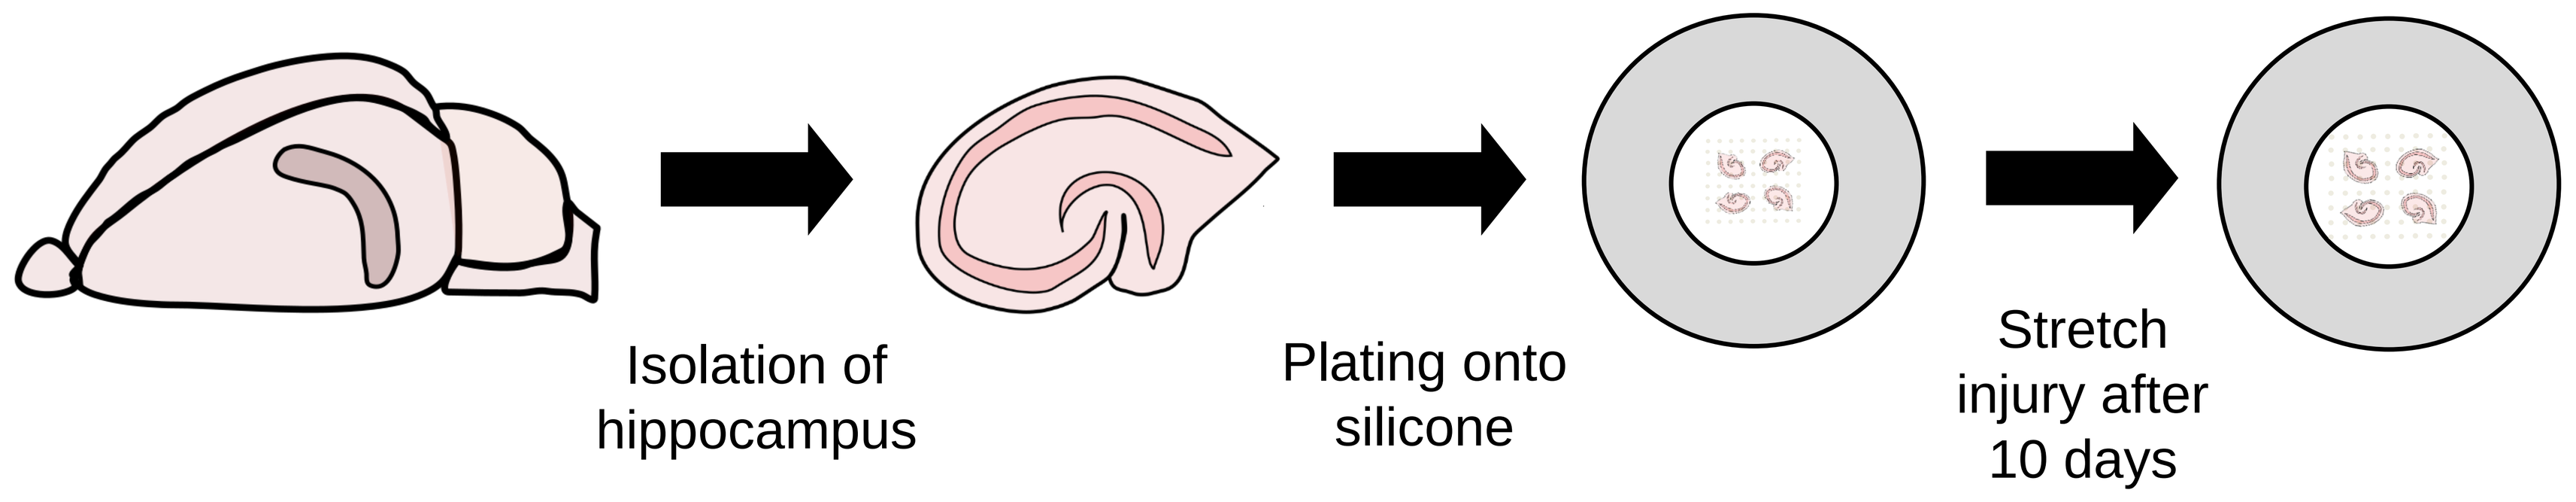
Isolation of hippocampus
Plating onto silicone
Stretch injury after 10 days

- Hippocampi were removed from P8-10 Sprague-Dawley rats and sliced into 400µm sections. OHSCs were placed on deformable silicone membranes in custom-made stainless steel wells.

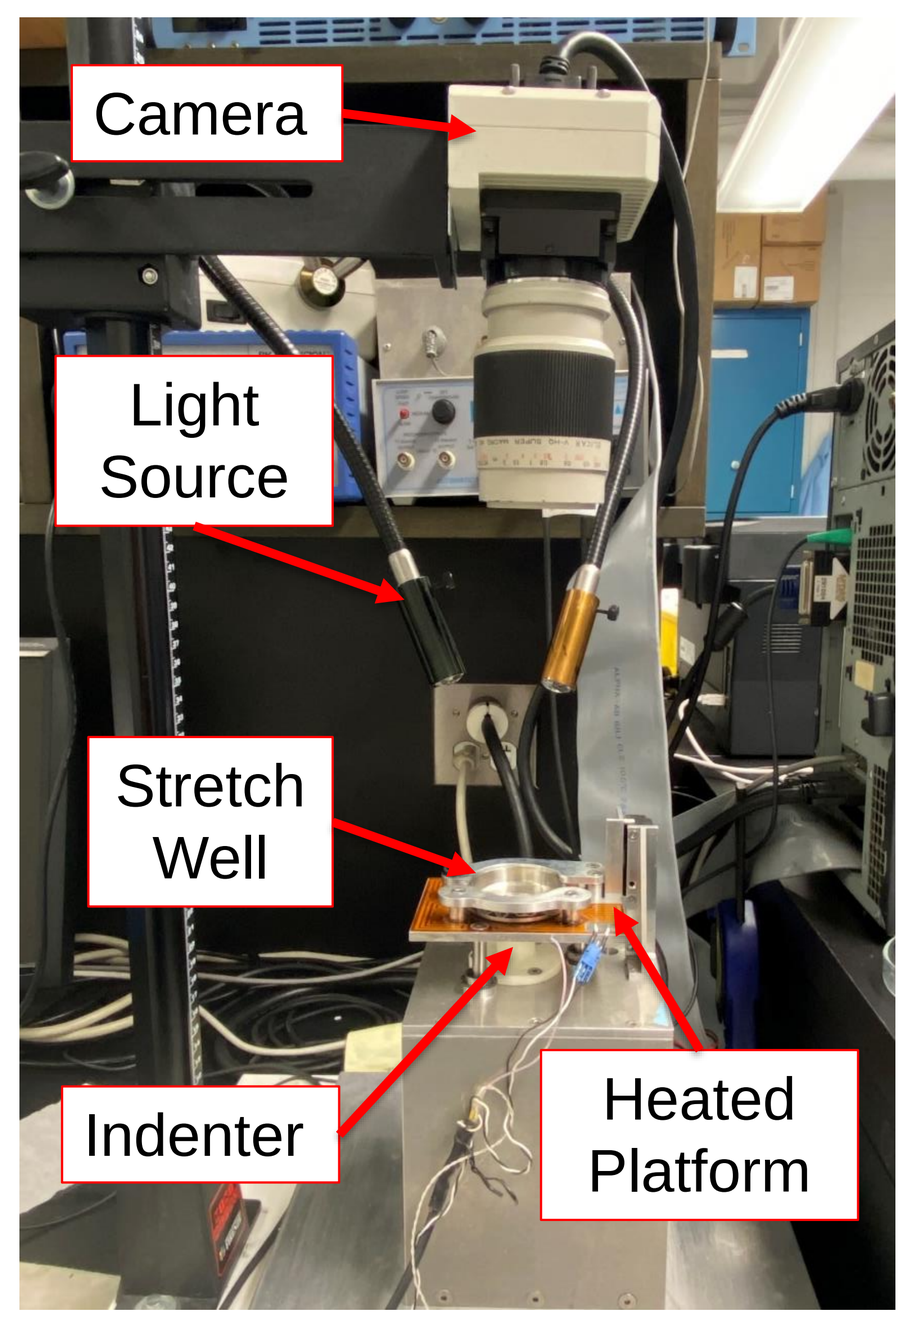
Camera
Light Source
Stretch Well
Indenter
Heated Platform

- Every 2-3 days half the conditioned media was replaced with fresh media. The OHSCs were rocked in an incubator at 37ºC with 5% CO_2.
- In order to increase the strain achieved by these OHSCs, the silicone membrane was patterned with a laser cutter to increase contact area.
- After at least 10 days *in vitro*, OHSCs were imaged with propidium iodide (PI) to confirm health.
- Healthy OHSCs, with <5% PI staining, were stretched and treated with NTS-105, progesterone, or vehicle (<0.003% DMSO) at one hour after injury. The average biaxial OHSC strain was verified with high-speed video to be 26% and the average strain rate was 9.8s^{-1}, modeling a moderate TBI.
- At 96 hours after injury, PI was used to measure cell death due to injury.

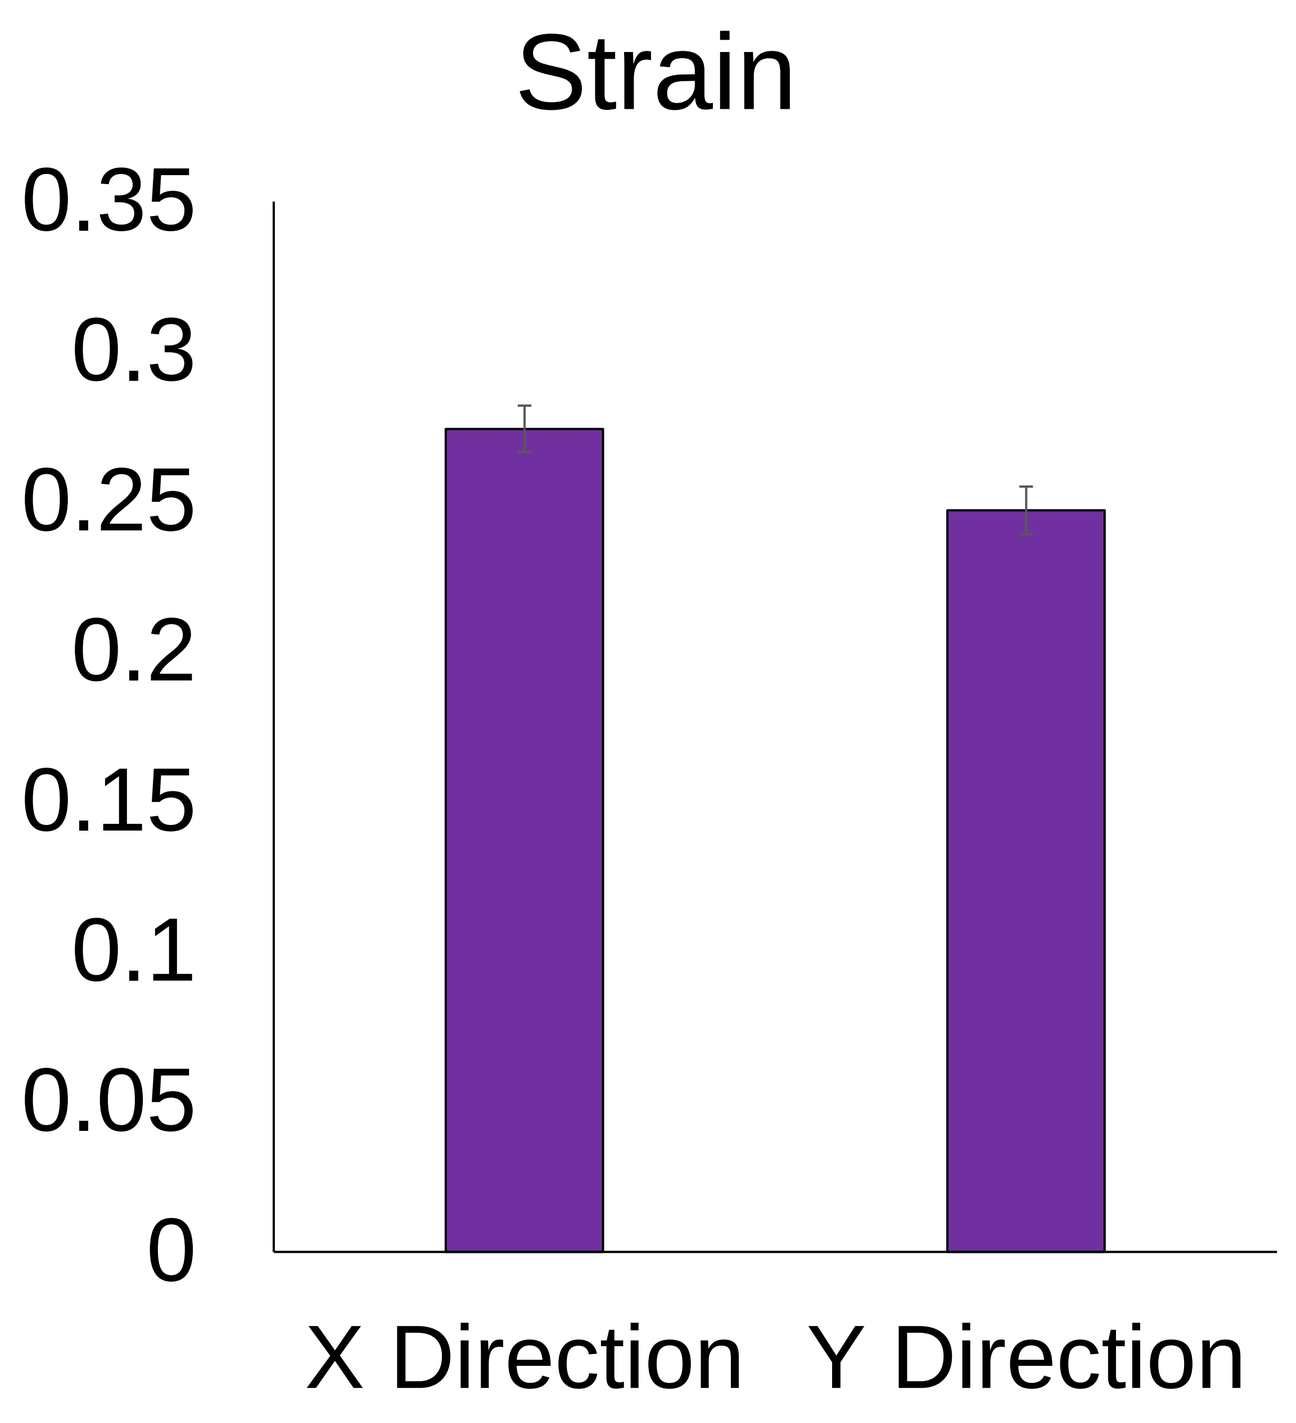
Strain
0.35
0.3
0.25
0.2
0.15
0.1
0.05
0
X Direction
Y Direction

Results

- NTS-105 prevented cell death at a wide range of concentrations (from 0.1nM to 30nM) at 96 hours after injury. Each group had N>=13 OHSCs. One-way ANOVA with Dunnett's test post-hoc. *=p<0.05; **=p<0.01; ***=p<0.001.

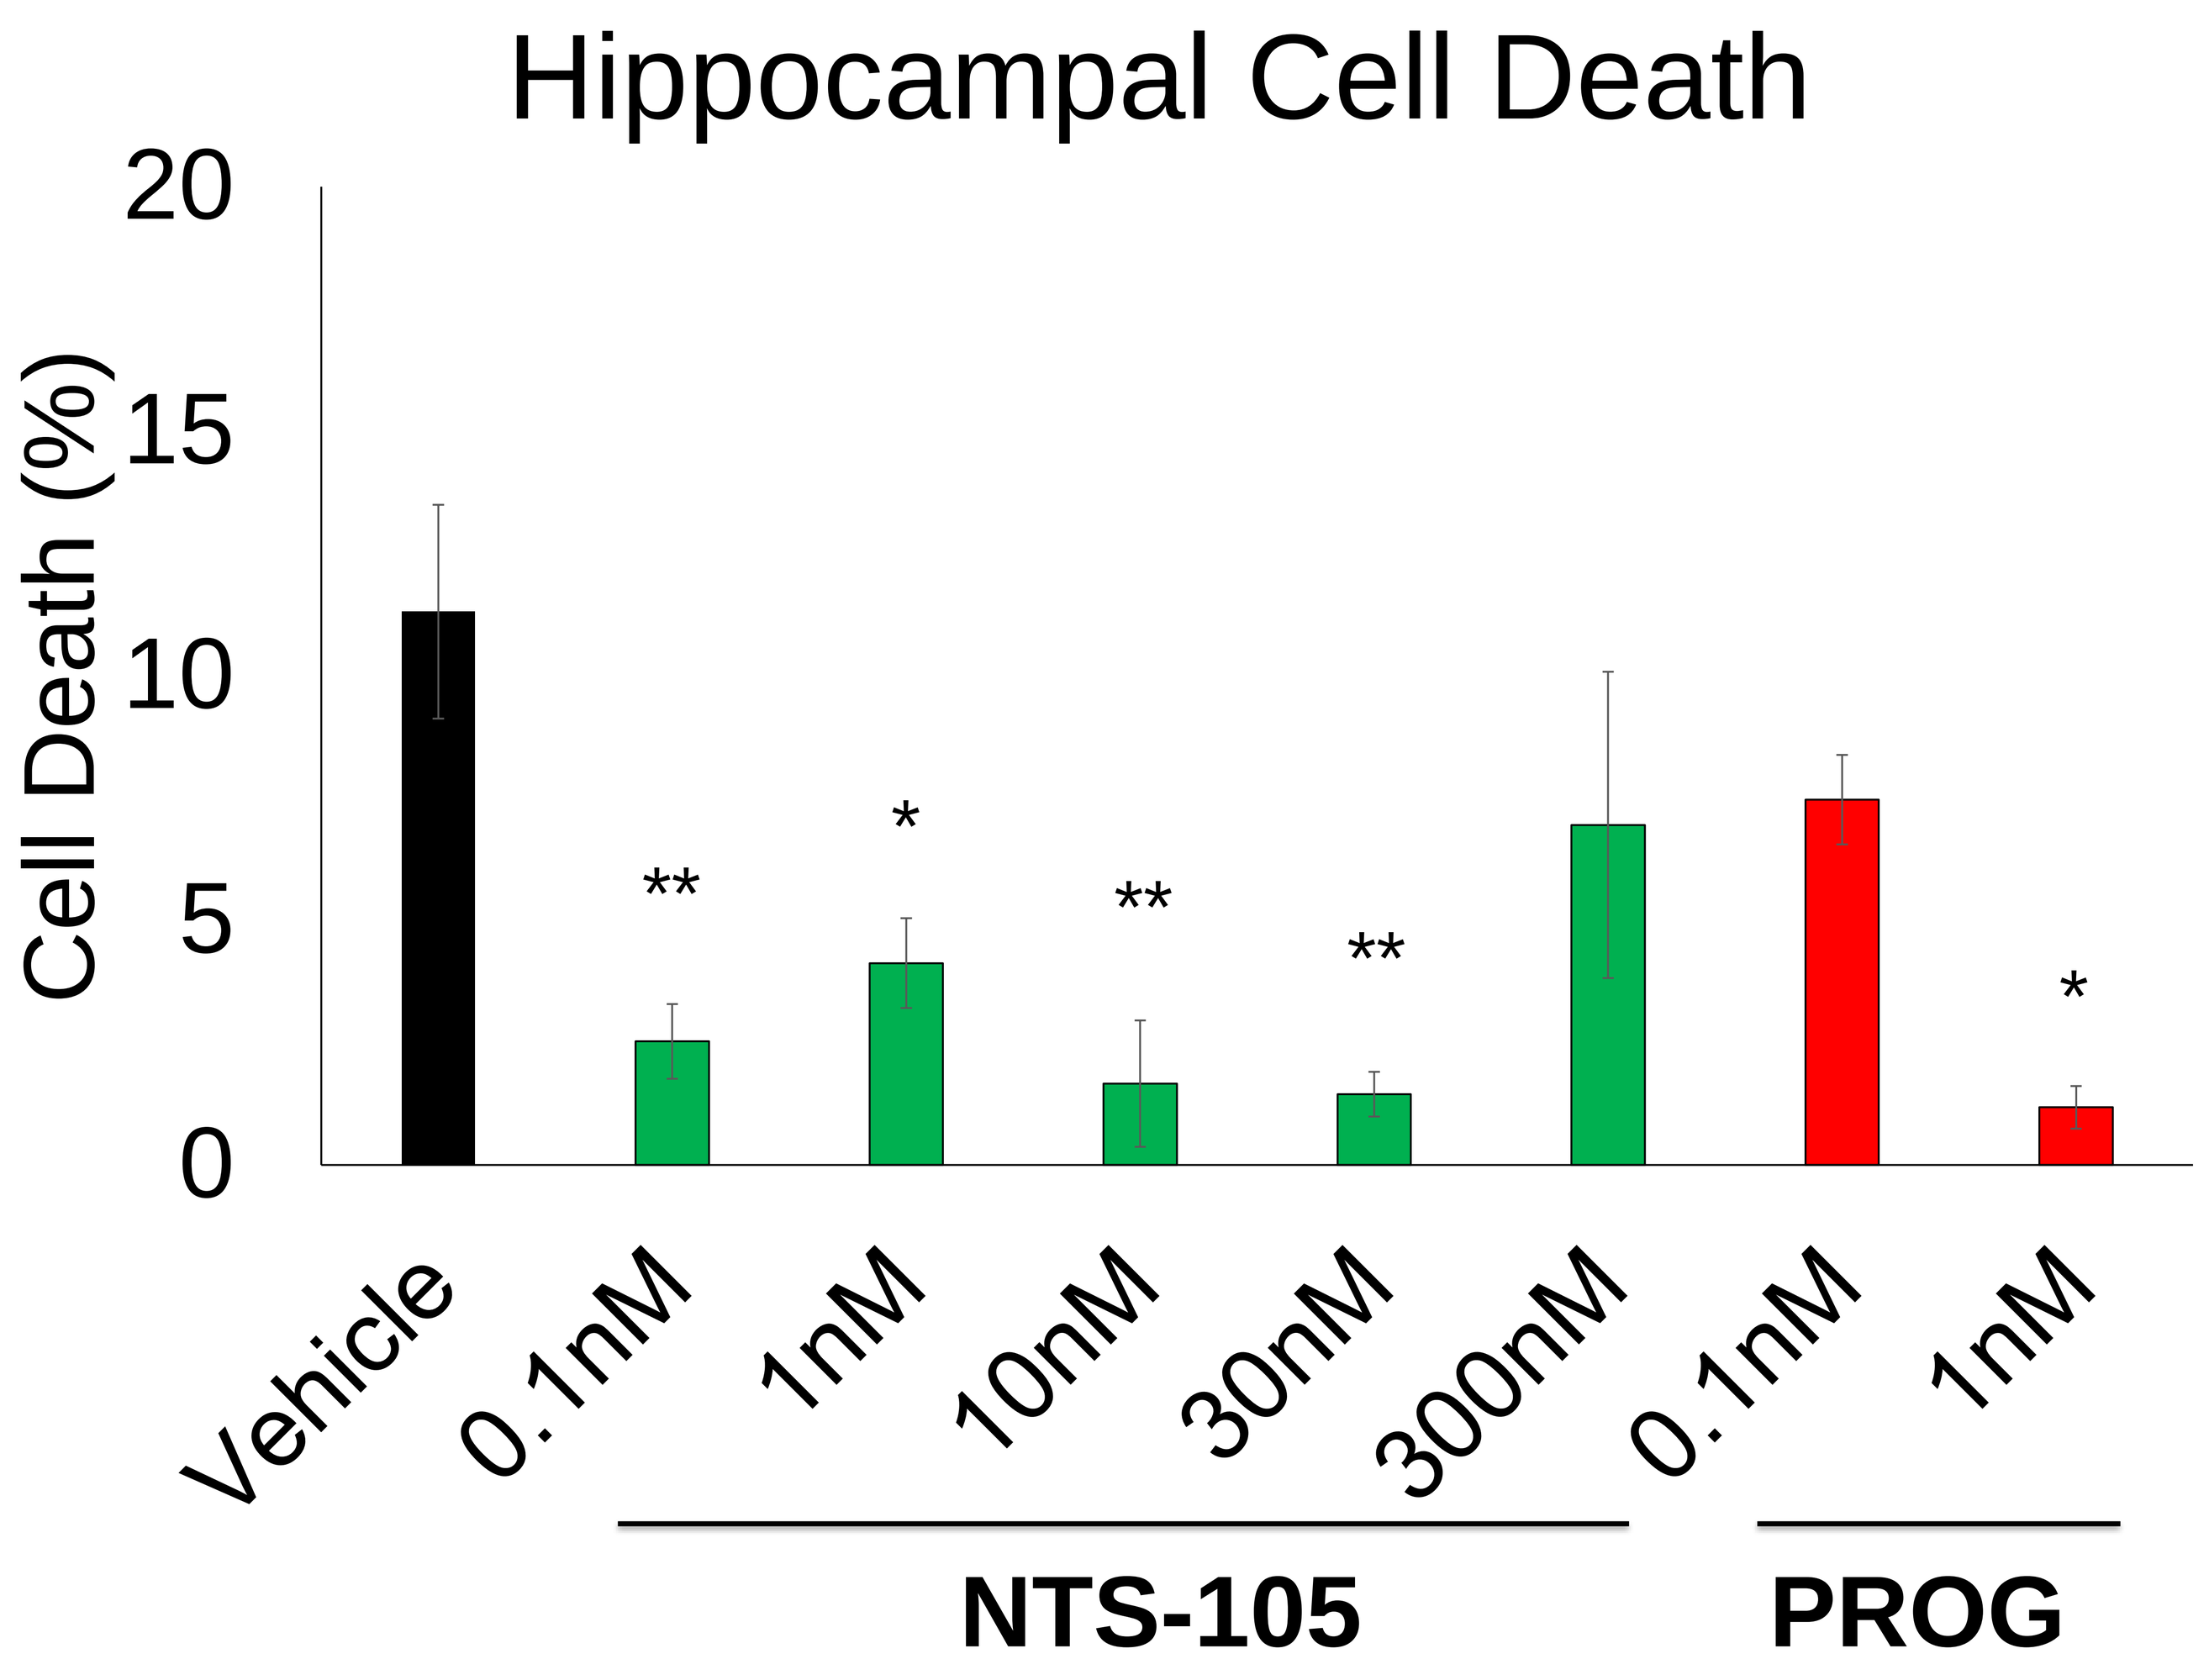
Hippocampal Cell Death
Cell Death (%)
20
15
10
5
0
**
*
**
**
*
Vehicle
0.1nM
1nM
10nM
30nM
300nM
0.1nM
1nM
NTS-105
PROG

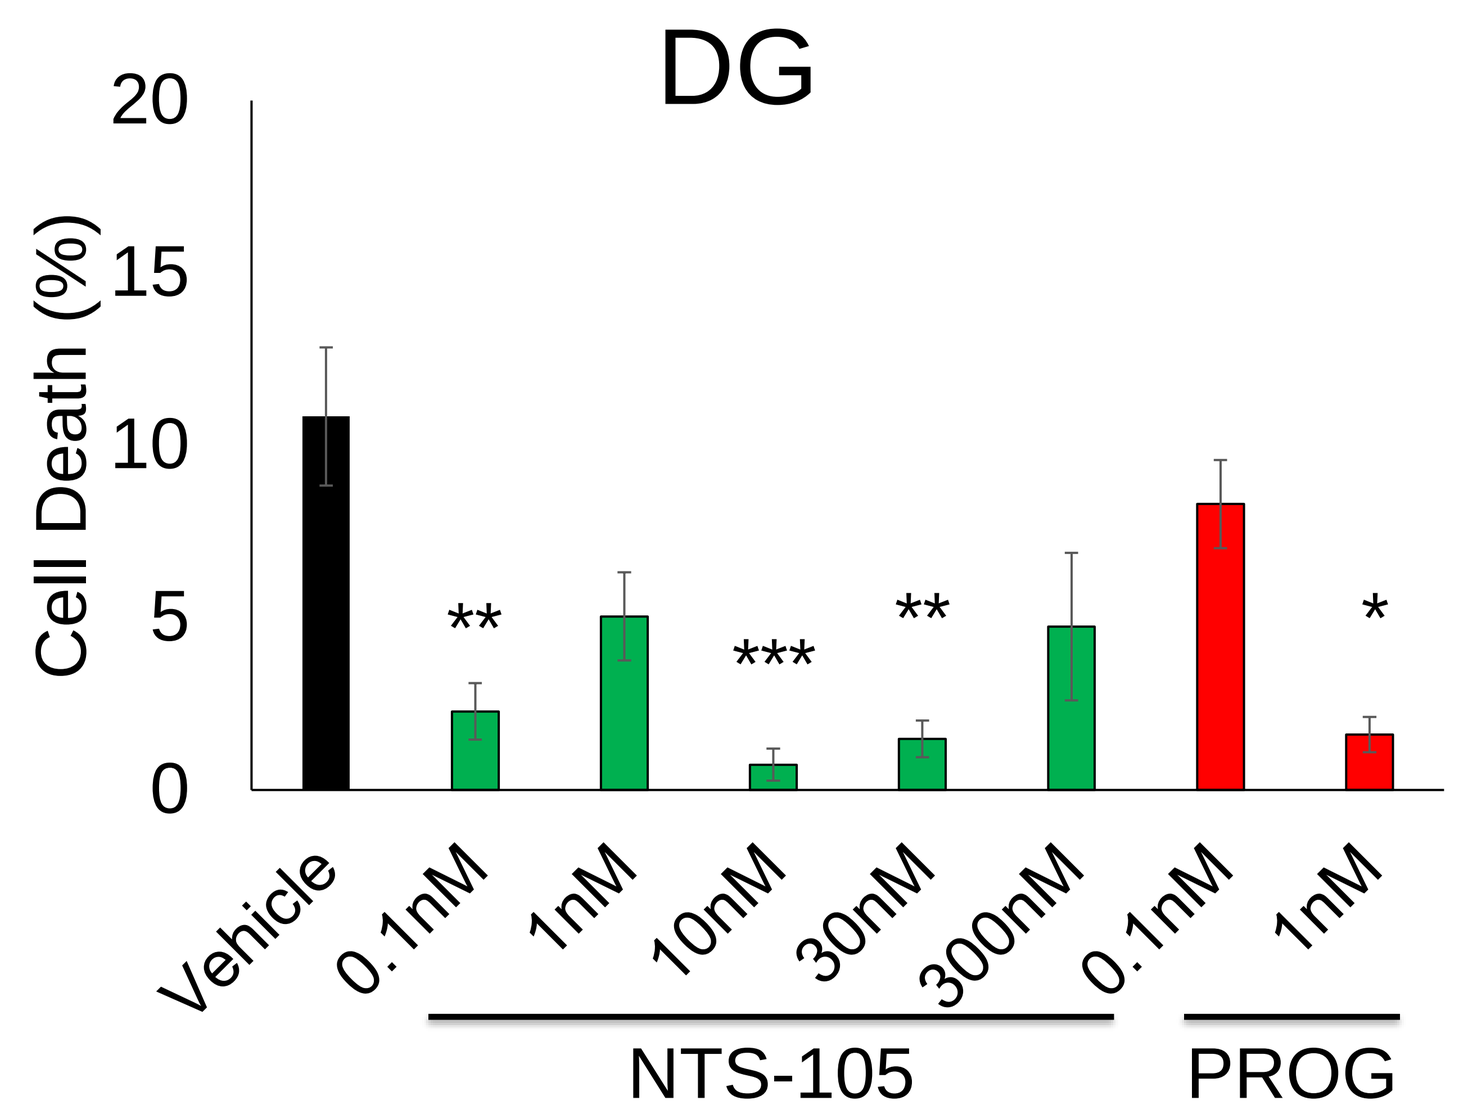
DG
Cell Death (%)
20
15
10
5
0
**

**
*
Vehicle
0.1nM
1nM
10nM
30nM
300nM
0.1nM
1nM
NTS-105
PROG

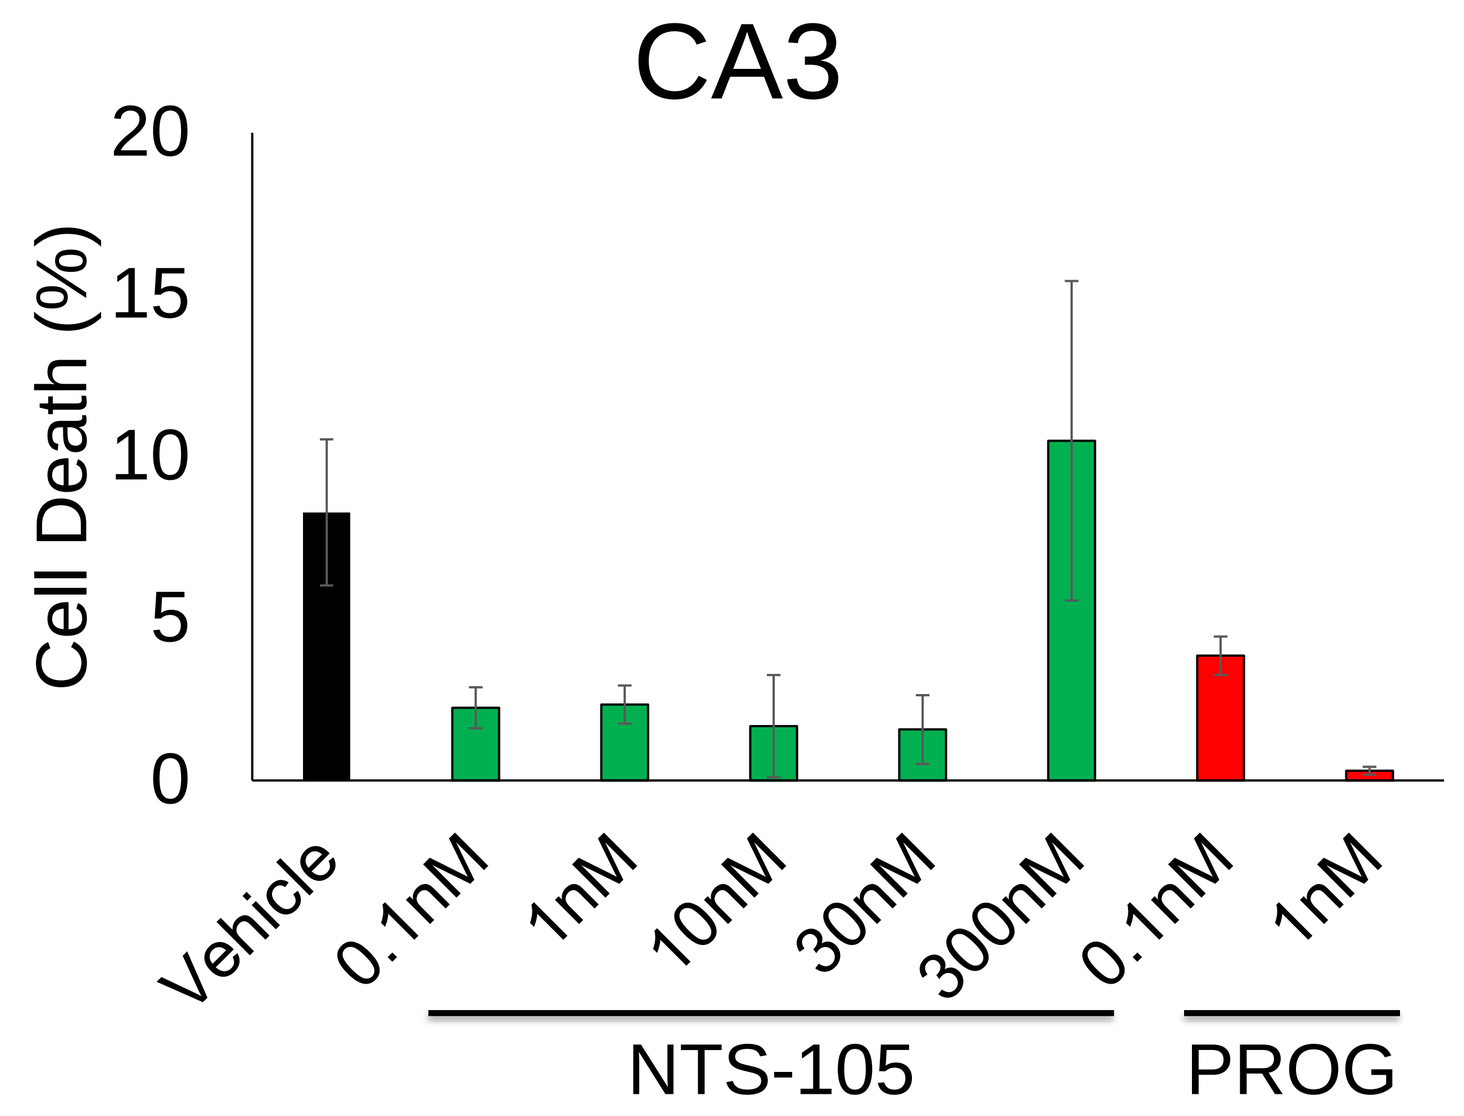
CA3
Cell Death (%)
20
15
10
5
0
Vehicle
0.1nM
1nM
10nM
30nM
300nM
0.1nM
1nM
NTS-105
PROG

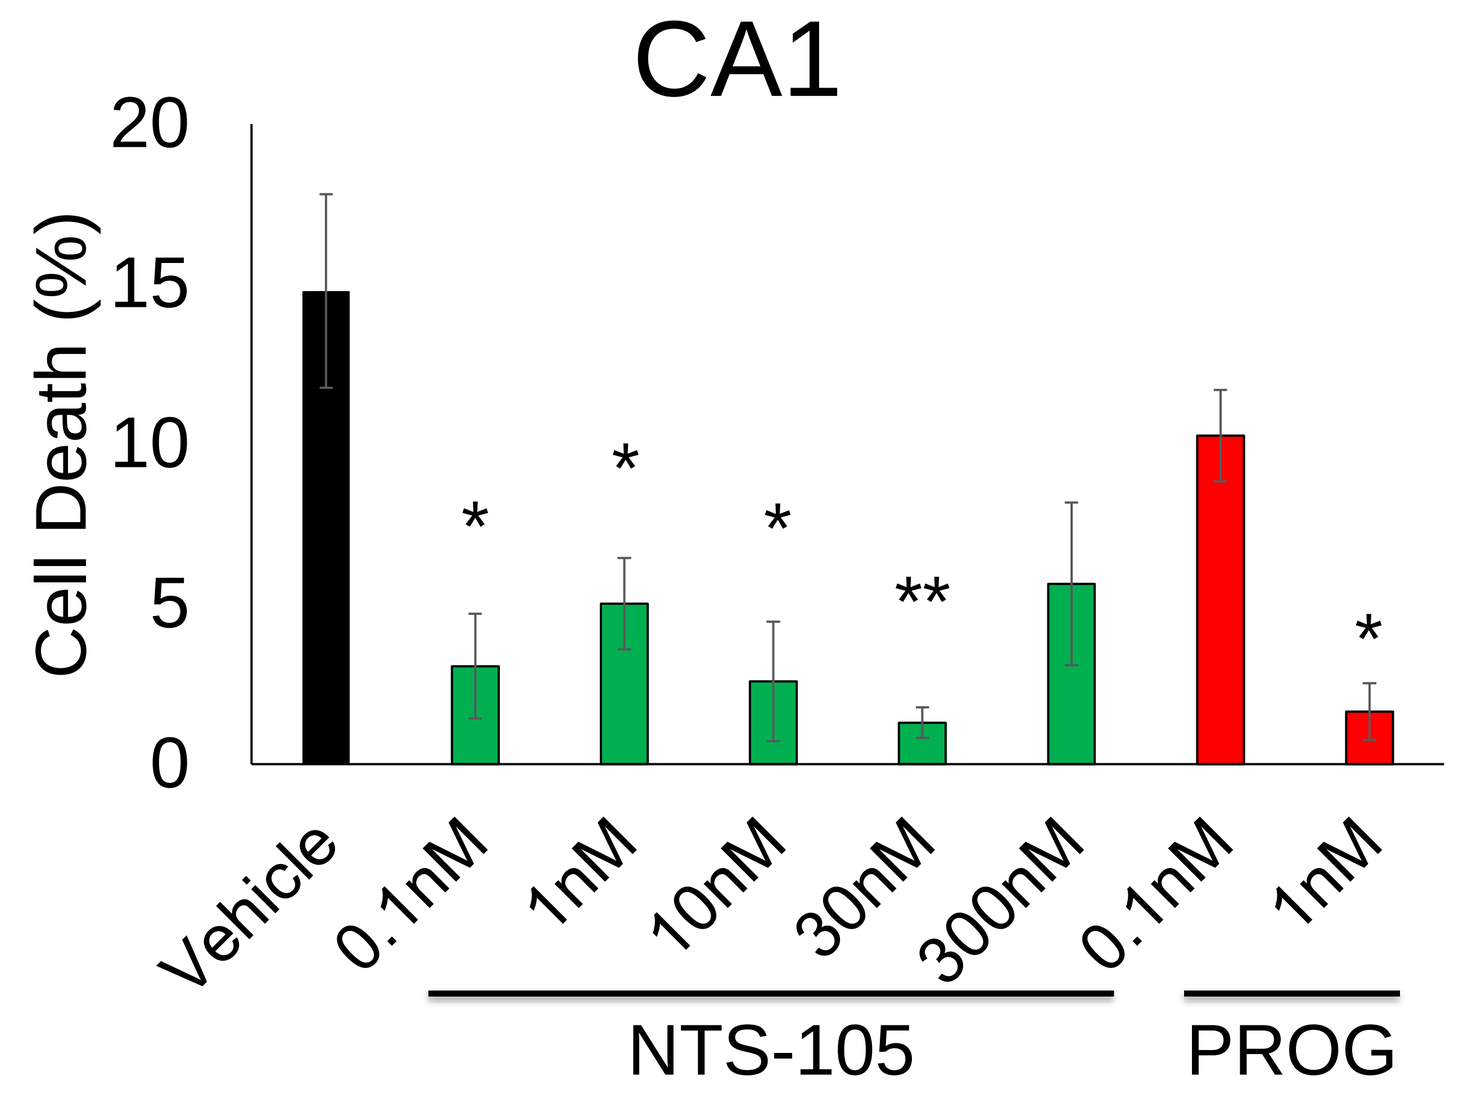
CA1
Cell Death (%)
20
15
10
5
0
*
*
*
**
*
Vehicle
0.1nM
1nM
10nM
30nM
300nM
0.1nM
1nM
NTS-105
PROG

- In the DG, 1nM progesterone and several concentrations of NTS-105 were neuroprotective.
- In the CA3, due to the low cell death in the vehicle group, no post-hoc comparisons were significant.
- In the CA1, 1nM progesterone and NTS-105 concentrations between 0.1nM and 30nM were significantly neuroprotective.

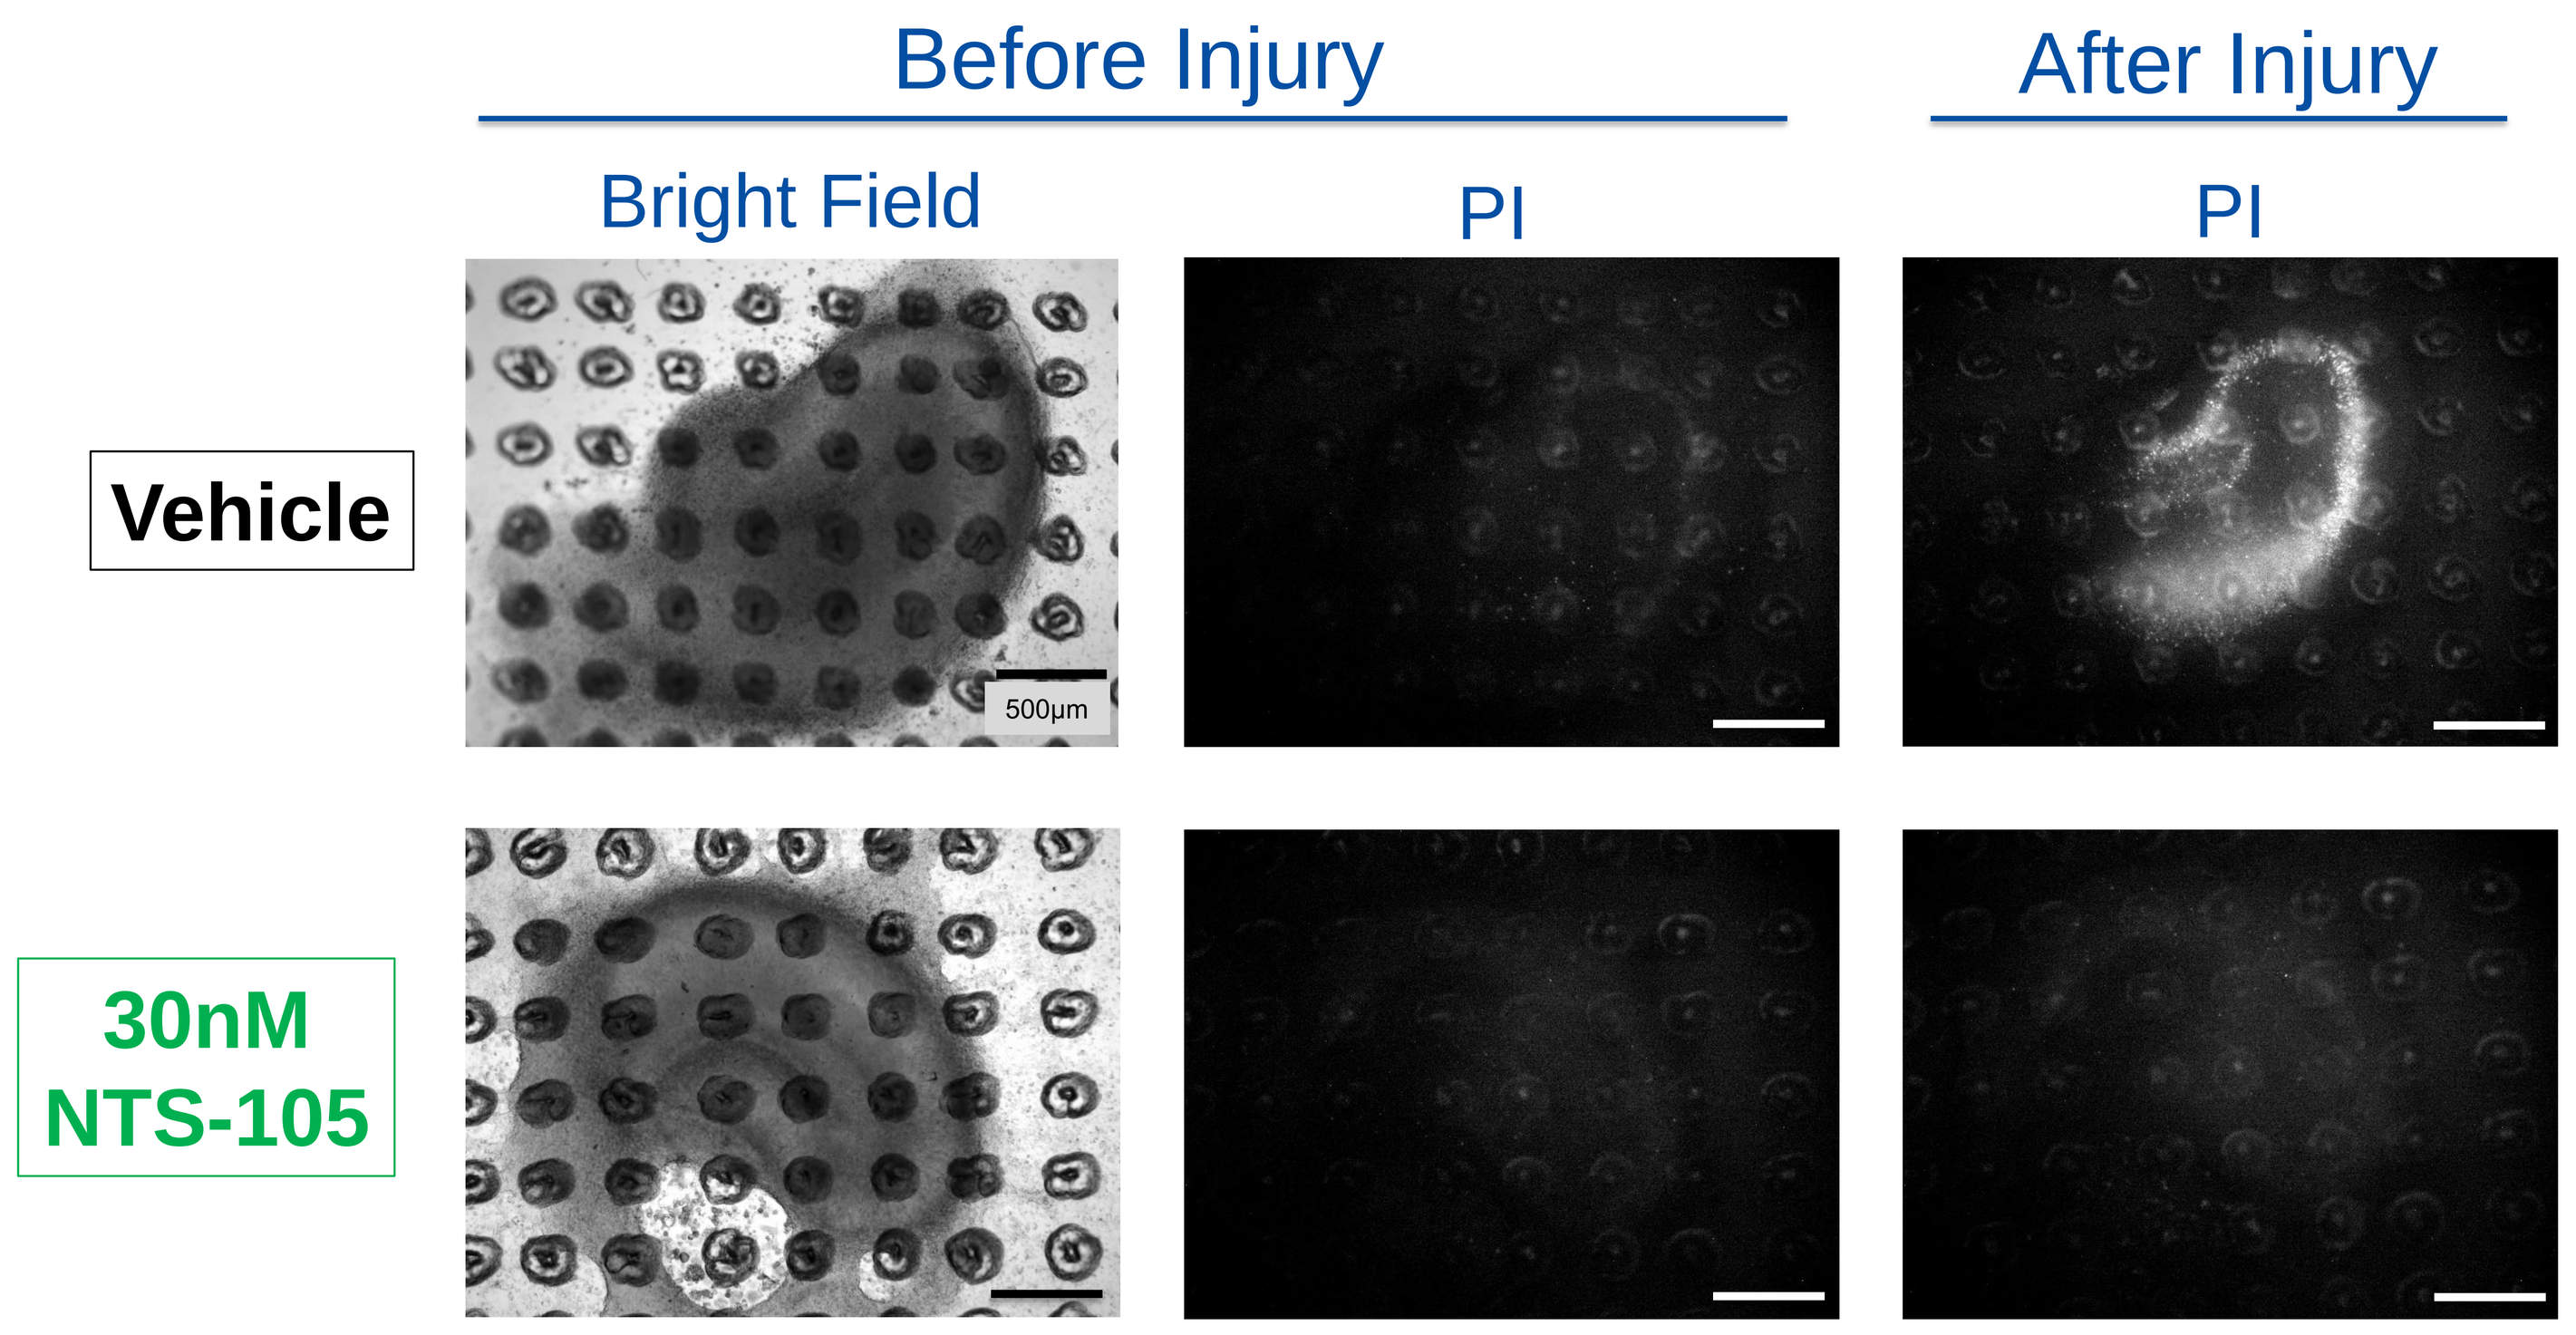
Before Injury
After Injury
Bright Field
PI
PI
Vehicle
500µm
30nM NTS-105

Discussion

- NTS-105 is able to reduce cell death at a wide range of concentrations in this model. 300nM NTS-105 was not as neuroprotective as lower concentrations. Like progesterone, NTS-105 may have a U-shaped dose response curve.
- Progesterone can be difficult to dissolve in water making it challenging to deliver clinically [4]. A neuroactive steroid with higher water solubility, like NTS-105, that is also neuroprotective may serve as a better option.
- Clinical trials of progesterone tested very few doses and they may have been too high [2]. The therapeutic window of NTS-105 in this model, from 0.1 to 30nM, can inform future *in vivo* treatment.

Future Work

- Interestingly, NTS-105 at a concentration of 0.1nM may reduce cell death further than 1nM. Future work may determine if this indicates a separate mechanism of action at this concentration.
- Identifying the targets of NTS-105 that protect against cell death. Starting with testing RU-486 (a progesterone receptor antagonist).
- Characterizing the inflammatory response in our model after stretch injury. Testing if NTS-105 is able to reduce pro-inflammatory cytokines.
- Testing the effects of NTS-105 on functional deficits, including electrophysiological activity, after injury.

Acknowledgements

This work was supported by NeuroTrauma Sciences, LLC.

References

[1] Centers for Disease Control and Prevention. Surveillance Report of TBI. U.S. Department of Health and Human Services. 2019.
[2] Donald Stein. Brain Inj. 2015.
[3] Gwen B Effgen and Barclay Morrison, III. J Neurotrauma. 2017.
[4] Iqbal Sayeed et al. Neuropharmacology. 2019.